Lack of Dose Flexibility in Solid Oral Controlled-Release Dosage Forms

G.A. Green, C. Berg, and N. Valdez
Accu-Break Pharmaceuticals, Inc., Florida, United States

ABSTRACT SUMMARY

Controlled-release (CR) medications offer many clinical and convenience advantages for patients as compared to immediate-release (IR) formulations. Among these are reductions of fluctuations in drug concentration and adverse side effects, increased compliance and a more convenient dosing regimen. The drawback however is the lack of dose flexibility. Many solid oral dosage CR medications are unsuitable for splitting due to the risk of absorption of a large amount of drug into the bloodstream if the drug delivery system is compromised. The aim of this study was to evaluate FDA-approved prescription solid oral CR medications on the US market with respect to the marketed dosage form and the labeled dosing instructions for the purpose of evaluating dosage form flexibility. Seventy-three reference-listed CR medications were identified (CR Products) that are marketed as solid oral dosage forms. Fifty-one (70%) have titration recommended as part of their dosing instruction; of these 28 (55%) are unscored tablets, 18 (35%) are capsules, and 5 (10%) are scored tablets. Of the latter, 4 of 5 are suitable for cutting according to the manufacturer's instructions. In other words, 47 out of 51 (92%) of the CR Products for which titration is recommended per the labeled dosing instructions are supplied in a dosage form that is unsuitable for splitting. This lack of dosage form flexibility can lead to an inefficient use of healthcare resources and poor patient compliance. Novel tablet technologies are available which may address this problem.

INTRODUCTION AND BACKGROUND

Repeated administration of a drug at regular intervals may maintain continuous therapeutic action over an extended period of time. The process can involve different combinations of dose and dose intervals, making it difficult for patients to adhere to the therapeutic regimen. The development of oral CR delivery systems stemmed from the need to reduce the dosing burden by releasing active substance at a passively controlled rate over an extended period of time. The advantages include reductions in fluctuations in drug concentration and adverse side effects, increases in patient comfort and compliance, and, potentially, reduced healthcare costs.

Since the early 1970s, CR delivery systems exploded in popularity. This led to the use of drug delivery technologies as a life cycle management tool and re-patentability strategy employed by many pharmaceutical manufacturers for their leading brands. The rapid and continuous innovation behind CR technologies is impressive, as evidenced by the creation of specialty drug delivery companies. However, the main drawback of CR products has, ironically, become their lack of dose flexibility. CR systems employ a variety of technologies including matrix systems, slowly eroding devices, osmotic-controlled systems and various polymer coatings. CR products also usually contain a greater amount of drug than the IR versions. Thus, they are typically unsuitable for breaking, crushing or masticating, as doing so may result in the releasing of a dangerously large amount of drug into the bloodstream. Several published studies comparing the in Vitro dissolution and in Vivo pharmacokinetics of whole and half CR theophylline tablets indicate that half tablets dissolve faster and result in higher plasma levels.

Thus, for CR medications, if titration or a change of dose is recommended in the product's dosing instructions, dose adjustment can be somewhat more onerous as a result of the risks associated with modifying the dosage form. Conversely, titration with IR drugs often can be achieved by splitting a tablet. Nevertheless, even with IR tablets, there may be significant variations in drug content of the subdivided parts due to inaccurate breaking. Patients taking oral CR dosage forms are, in most cases, relegated to acquiring a new prescription(s) for a different strength(s) at the pharmacy, or taking multiple doses at one time. This can be time consuming and result in increased costs and reduced compliance. We sought to evaluate FDA-approved, prescription solid oral CR medications on the US market with respect to the marketed dosage form and the labeled dosing instructions for the purpose of identifying whether tablet splitting was an option for patients for adjusting doses.

EXPERIMENTAL METHODS

The FDA's Electronic Orange Book was used to search for approved CR medications listed as of December 31st, 2008. Using the "Search by Proprietary Names" function, the following search terms were inputted: "Controlled Release", "CR", "Extended Release", "ER", "Sustained Release", "SR", "Modified Release", "MR", "Time Release", "TR", "XL", "XR" and "HS". The results were filtered for capsules and tablets only, and the reference listed drugs (RLDs) were chosen. Any product identified using this methodology with a delivery mechanism intended specifically for release of drug within the colon was excluded from the database (e.g. Asacol; HalfLytely). The resulting database of oral solid CR RLDs are referred to as the "CR Products".

The prescribing information for each CR Product was obtained from the Physicians' Desk Reference (PDR; 1996 to 2008 versions) and the reference websites Epocrates Online and Drugs.com. The CR Products were characterized by dosage form (tablet or capsule) and presence or absence of a score mark. The Dosage and Administration instructions were reviewed to determine if titration was recommended as part of the treatment guidelines. Finally, each CR Product was reviewed for guidance as to whether it was appropriate or not to split the dosage form (e.g. cut in half to obtain a smaller dose).

RESULTS AND DISCUSSION

A total of 73 RLDs were identified according to the search methodology and criteria as having a CR formulation and supplied as an oral capsule or tablet ("CR Products"). The breakdown of these CR Products with respect to the dosage forms can be seen in Figure 1.

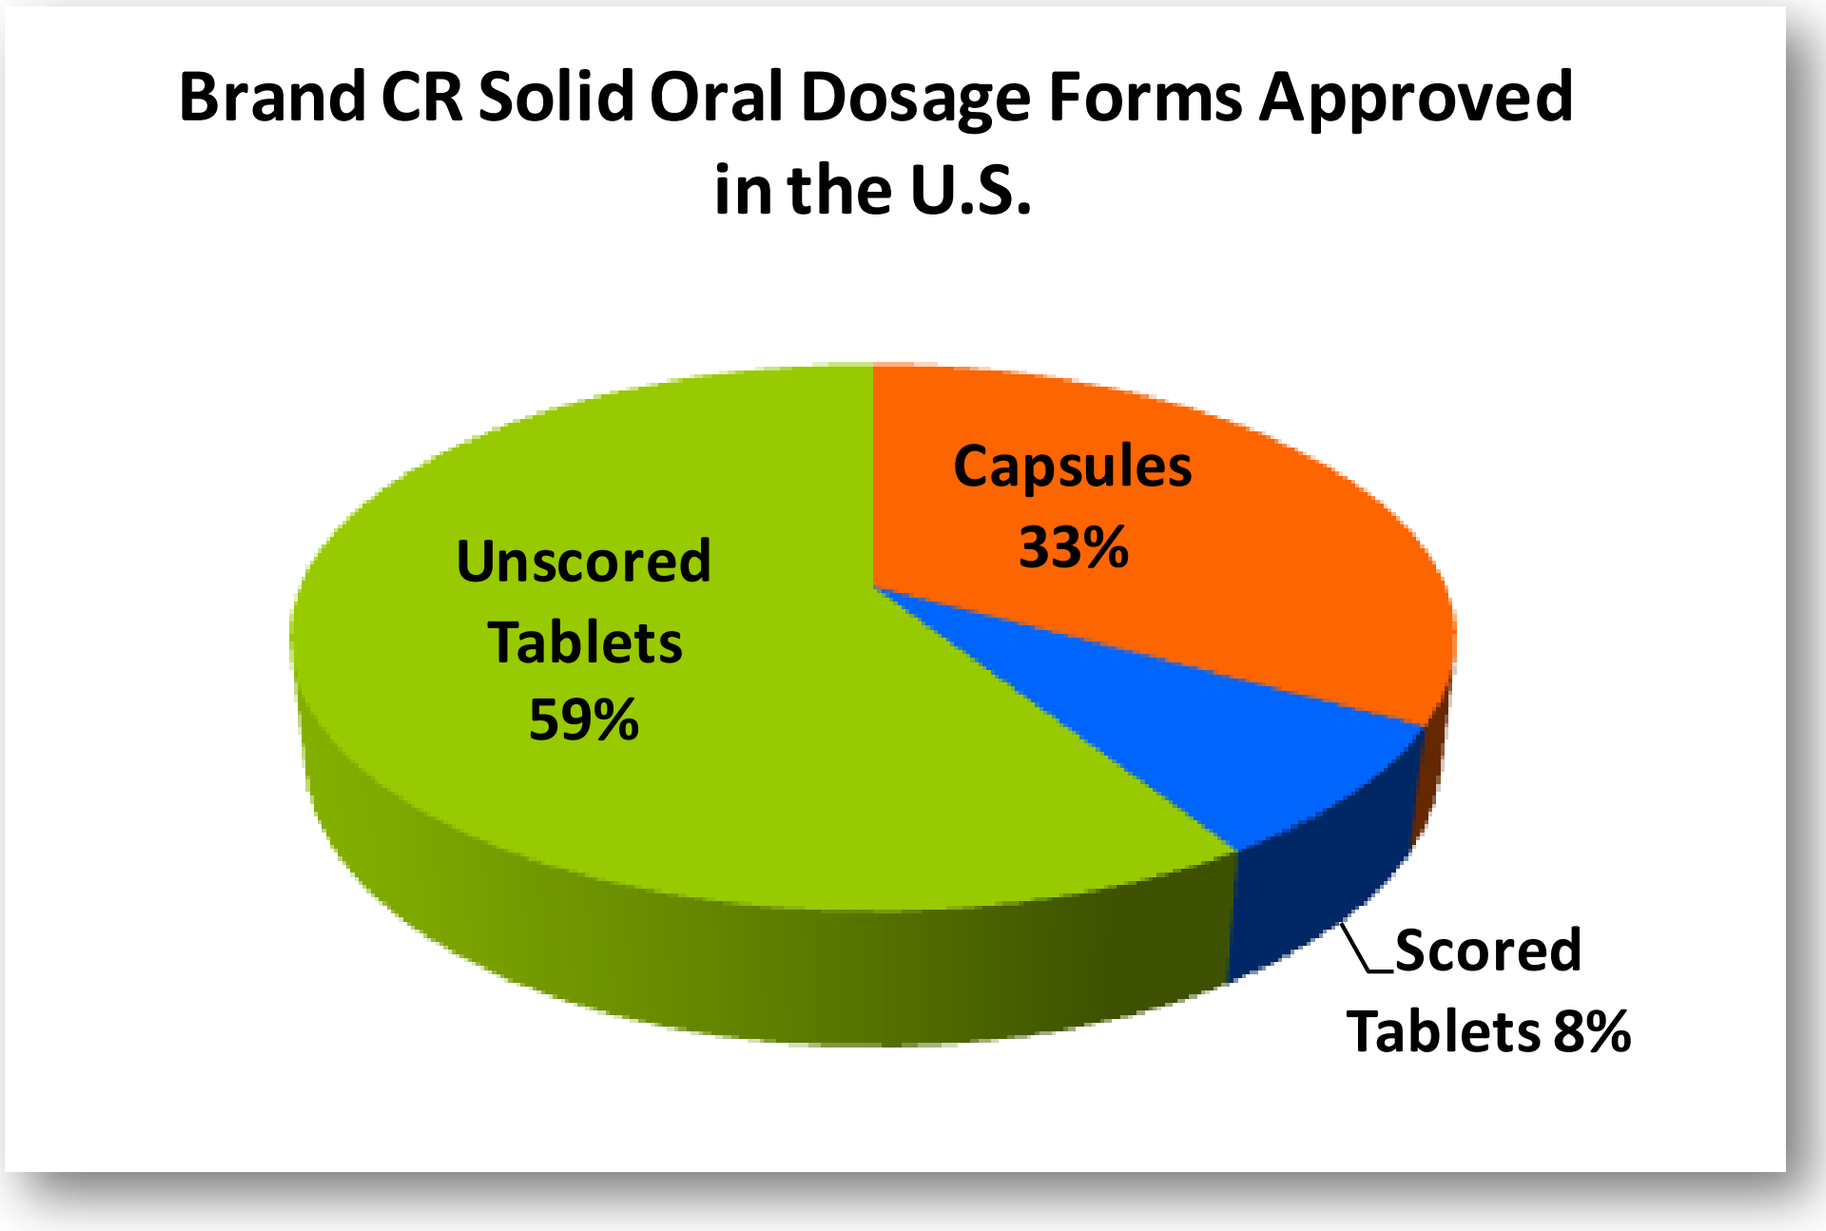
Brand CR Solid Oral Dosage Forms Approved in the U.S.
Capsules 33%
Unscored Tablets 59%
Scored Tablets 8%

Figure 1: The breakdown of the 73 FDA-approved (December 31, 2008) solid oral CR brand medications (reference listed drugs) by marketed dosage form.

Fifty-one (70%) CR Products have dosing instructions that include titration. (Figure 2) Of these 51, 28 (55%) are unscored tablets, 18 (35%) are capsules, and 5 (10%) are scored tablets. For the 5 CR Products that bear a score mark, which implies the tablet may be split, 4 have language stating they may be split into halves and one states that "[Product] tablets must be swallowed whole and never crushed or chewed." In other words, 47 (92%) of the CR Products with titration recommended are marketed in dosage forms that are not intended for subdivision.

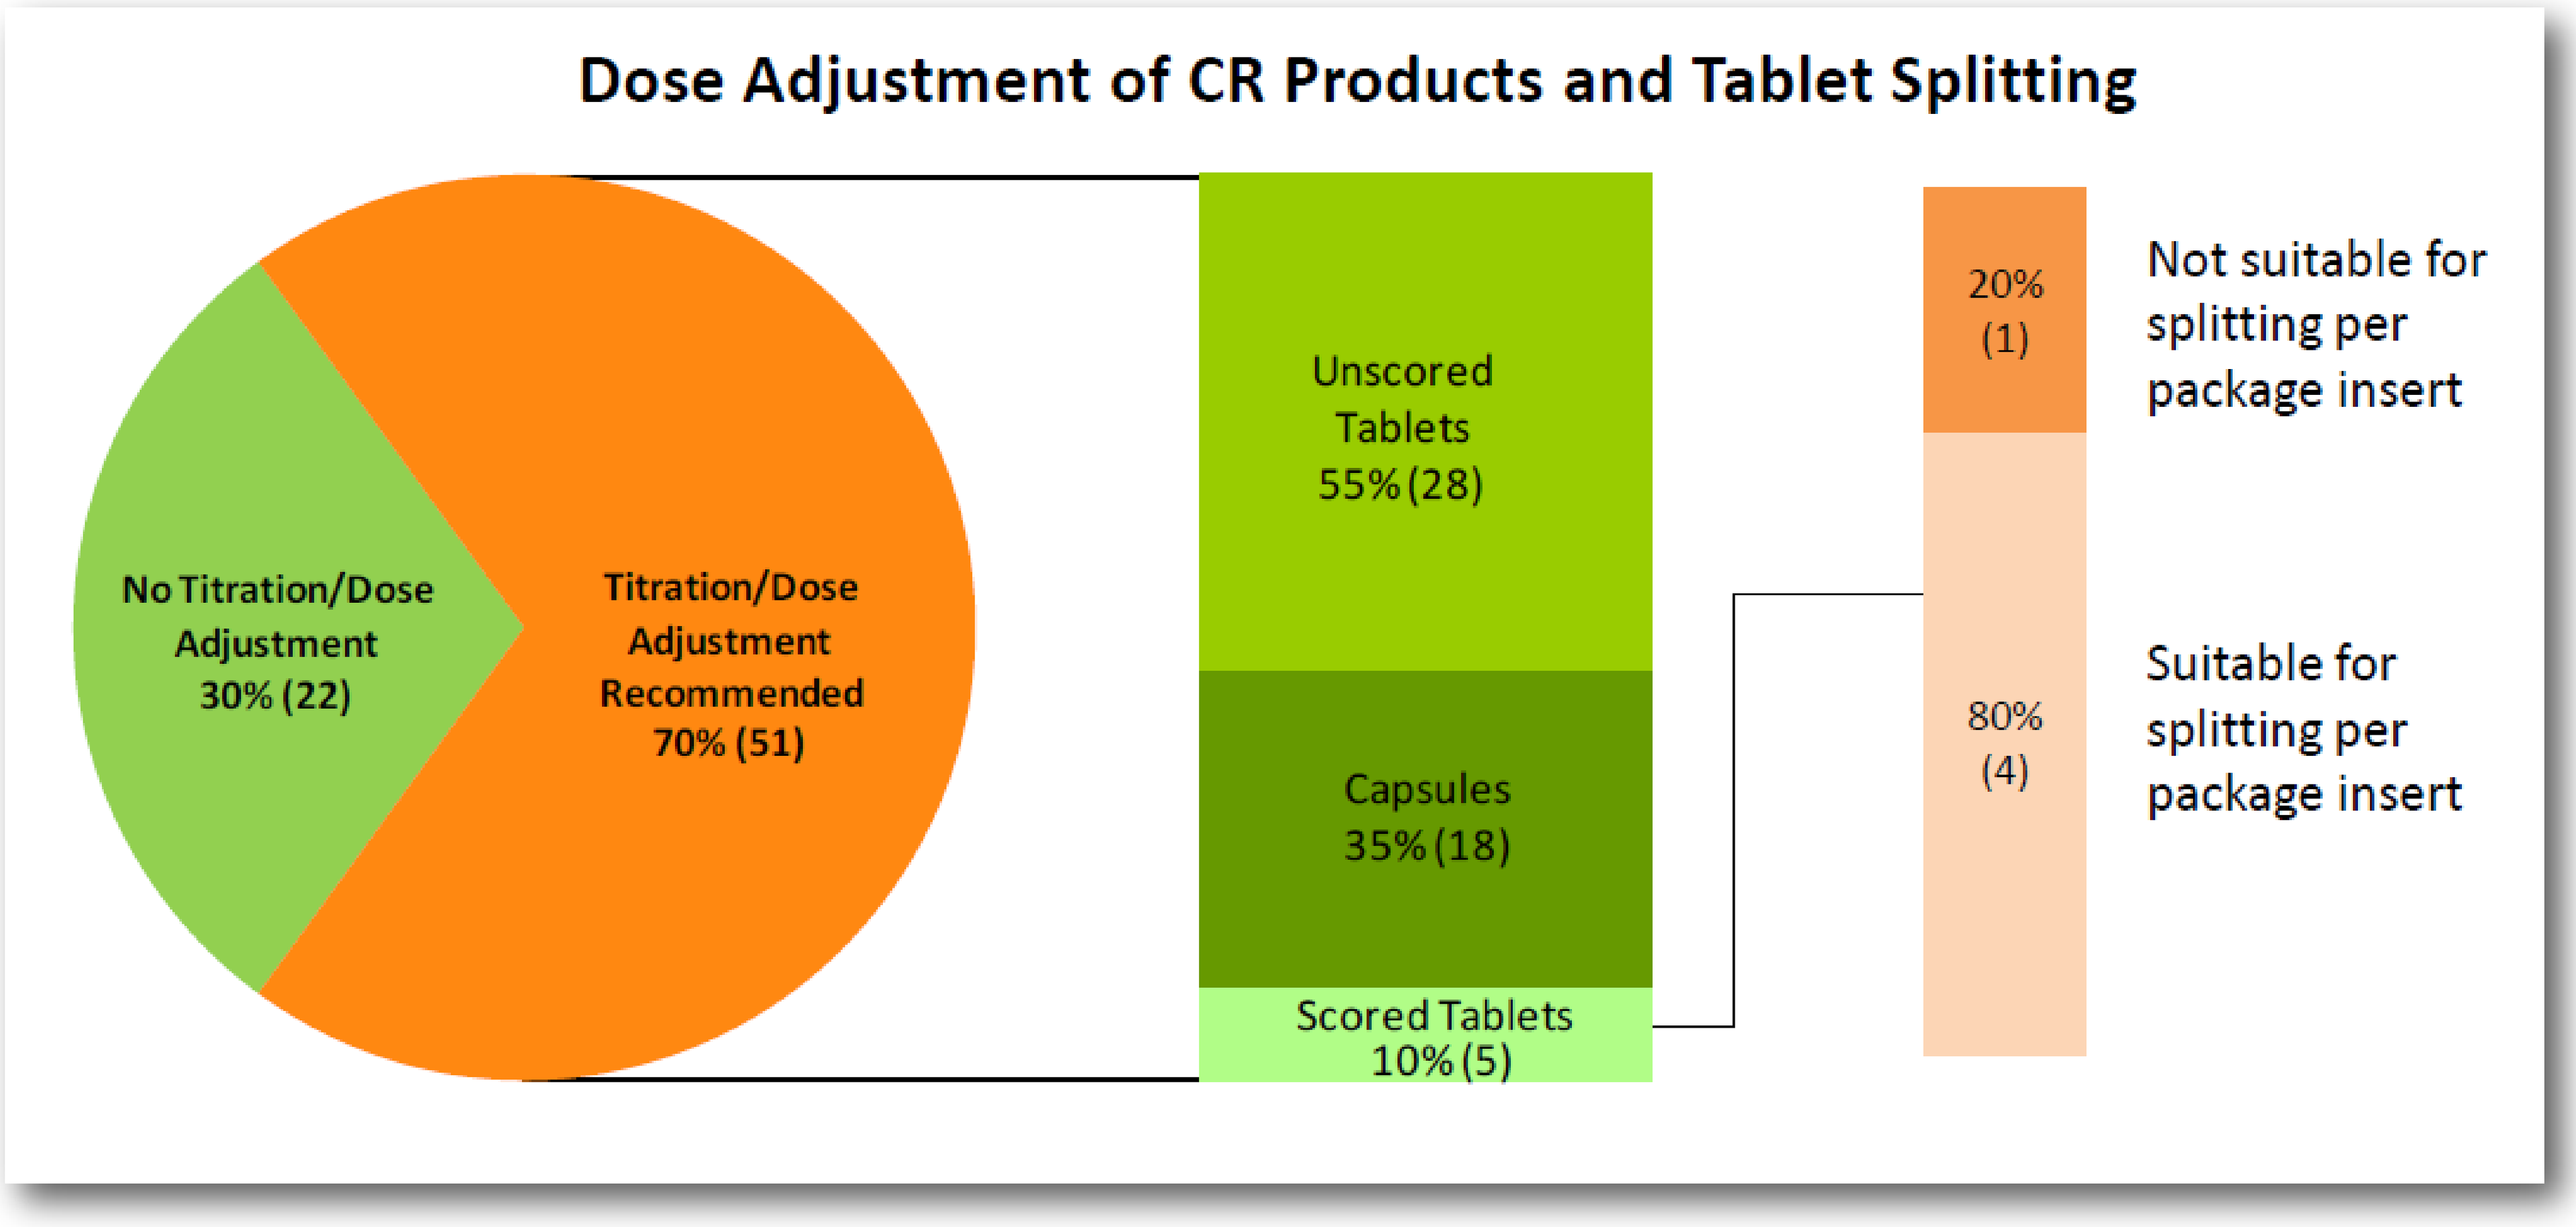
Dose Adjustment of CR Products and Tablet Splitting
No Titration/Dose Adjustment 30% (22)
Titration/Dose Adjustment Recommended 70% (51)
Unscored Tablets 55% (28)
Capsules 35% (18)
Scored Tablets 10% (5)
20% (1)
Not suitable for splitting per package insert
80% (4)
Suitable for splitting per package insert

Figure 2: The breakdown of the dosage forms for oral brand CR Products with language in the Dosage and Administration section of the package insert that suggests that dose adjustment is or may be necessary as part of the dosing regimen.

The results of the study suggest that for nearly all CR Products, dose adjustments per the labeled dosing instructions would necessitate either a new prescription, or having to swallow multiple doses. Certainly if the dose is decreased, there is but a single option which is obtaining a new prescription. Finding optimal therapeutic blood levels with CR medications is likely a trial and error process for most individuals as there have been reports of a lack of in Vitro-in Vivo correlation. Ideally, it would be beneficial for patients and caregivers to have the option of splitting CR Products into accurate smaller doses without significantly changing the in Vitro release profile. Our data shows that just 6% of the 73 CR Products can be split per the product package insert (Figure 3), making a change in dose for the rest of the CR Products inconvenient, wasteful and costly.

Figure 3: Percent of FDA-approved Prescription CR Products that are suitable for subdividing per the Package Inserts .

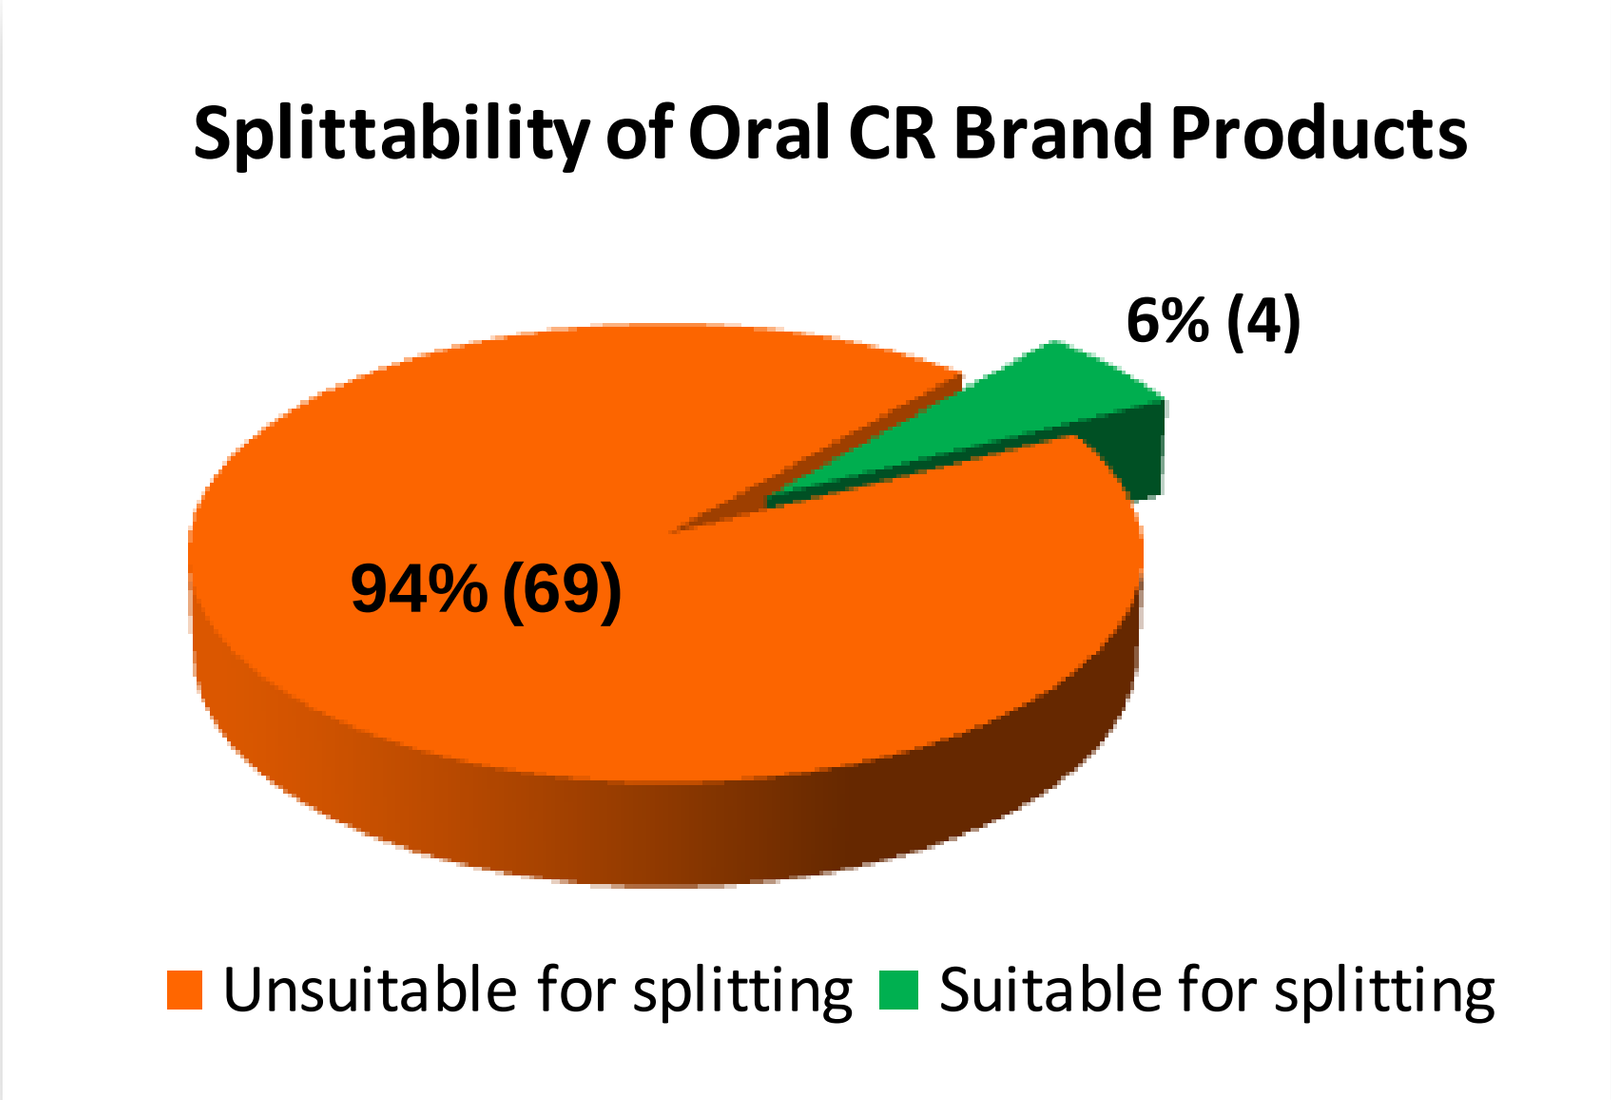
Splittability of Oral CR Brand Products
6% (4)
94% (69)
Unsuitable for splitting
Suitable for splitting

Two tablet technologies have been developed to address this specific problem. The first, Sanwa Kagaku Kenkyusho Co., Ltd., consists of divisible tablets that contain two enterically-coated cores which may be separated without affecting the release profile of the cores. The second technology, developed by Accu-Break Pharmaceuticals, Inc., utilizes a scored, drug-free break layer within each tablet to separate the CR dose into exact halves. (Figure 4) The Accu-Break technology allows the tablet to be split through the drug-free layer, resulting in two parts that contain exact half doses of the CR medication, each with the identical release kinetics as that of the whole tablet. This would allow patients to accurately titrate the CR medication by half doses without risking unpredictable doses or unintentionally higher drug plasma levels.

Figure 4: Accu-Break Technologies use a drug-free break layer within each tablet to segregate the dose into precise halves. Accu-Break CR tablets may be subdivided by hand into half doses without compromising the release kinetics, ensuring a safe and accurate partial dose of medication. (US Patent Nos. 7,318,935; 7,329,418; US Patent Pending 10/598,355.)

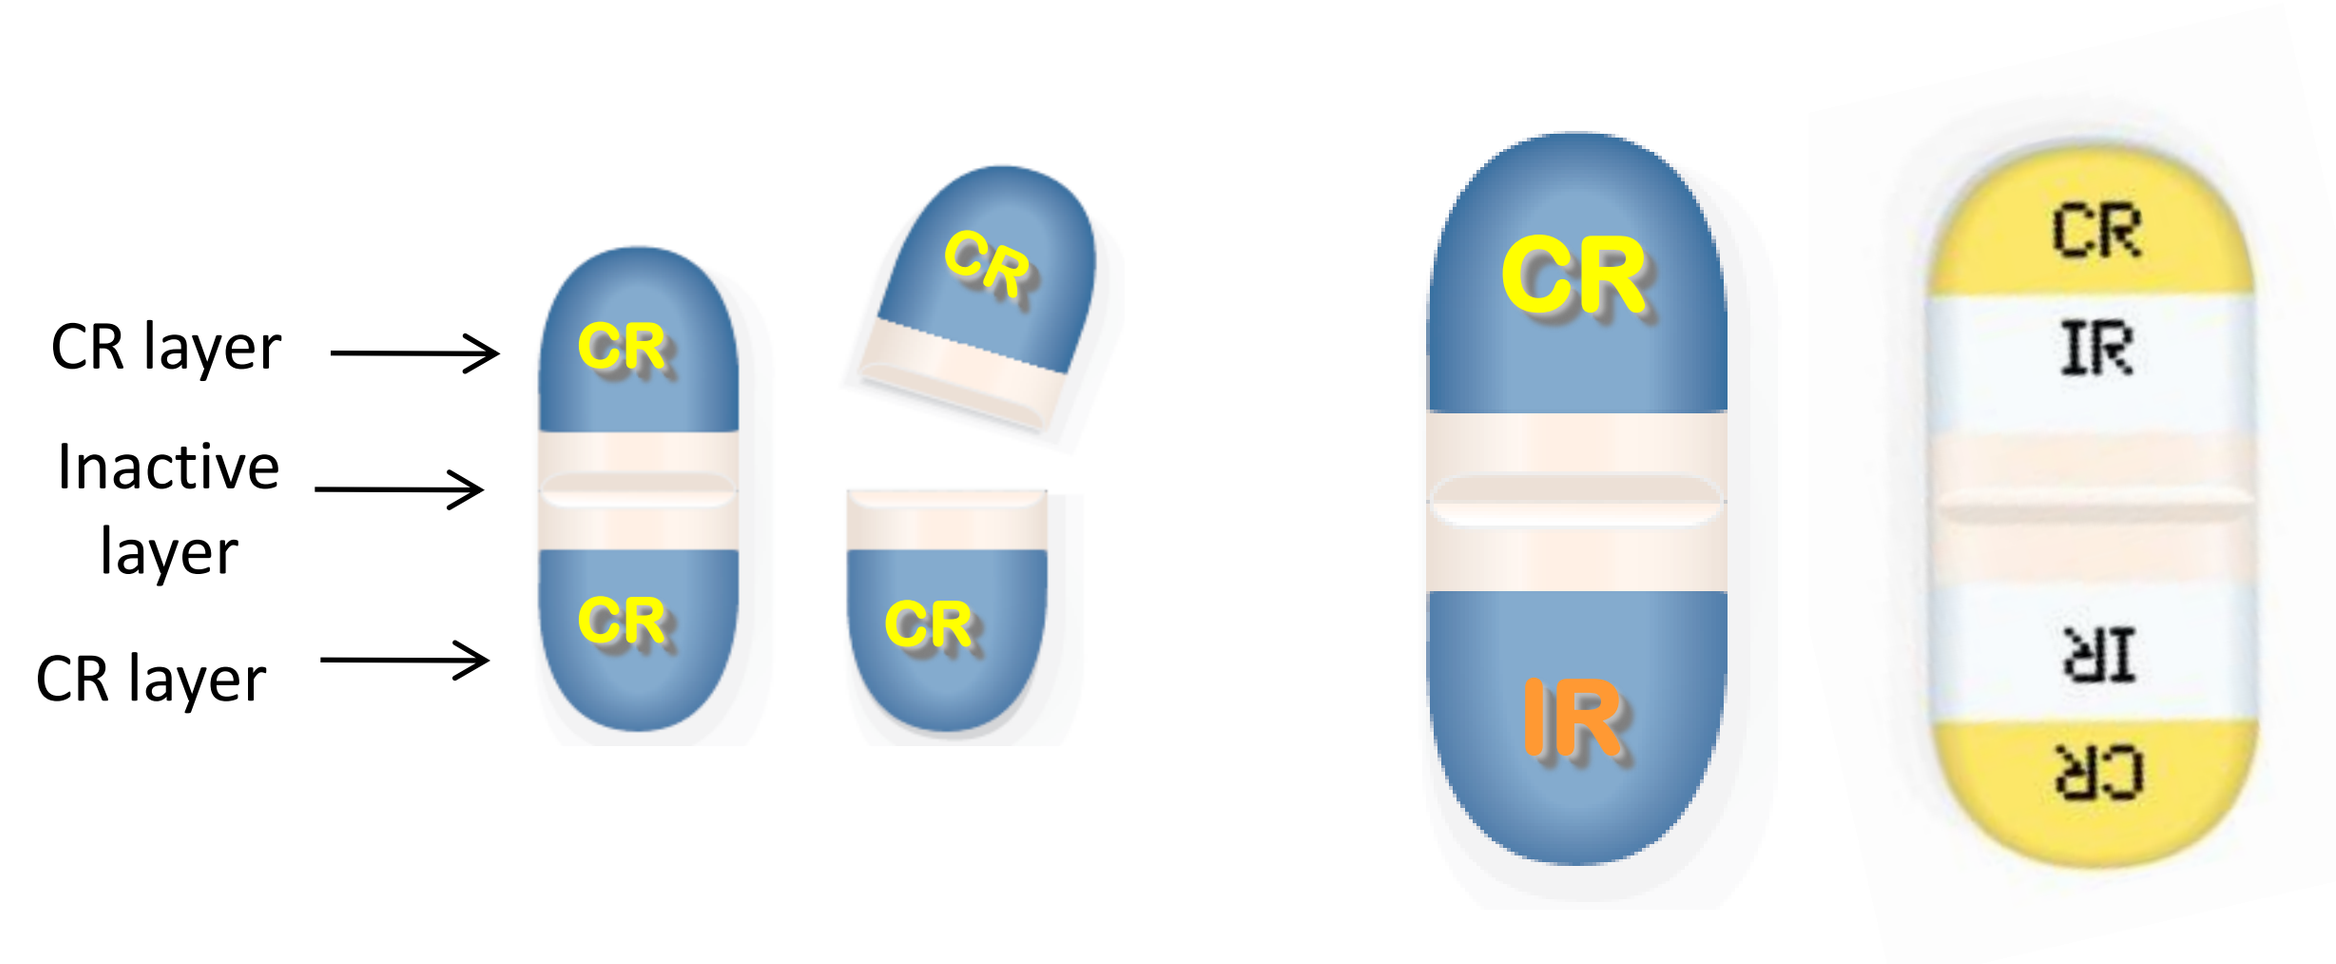
CR layer
Inactive layer
CR layer
CR
CR
CR
CR
CR
IR
CR
IR
IR
CR

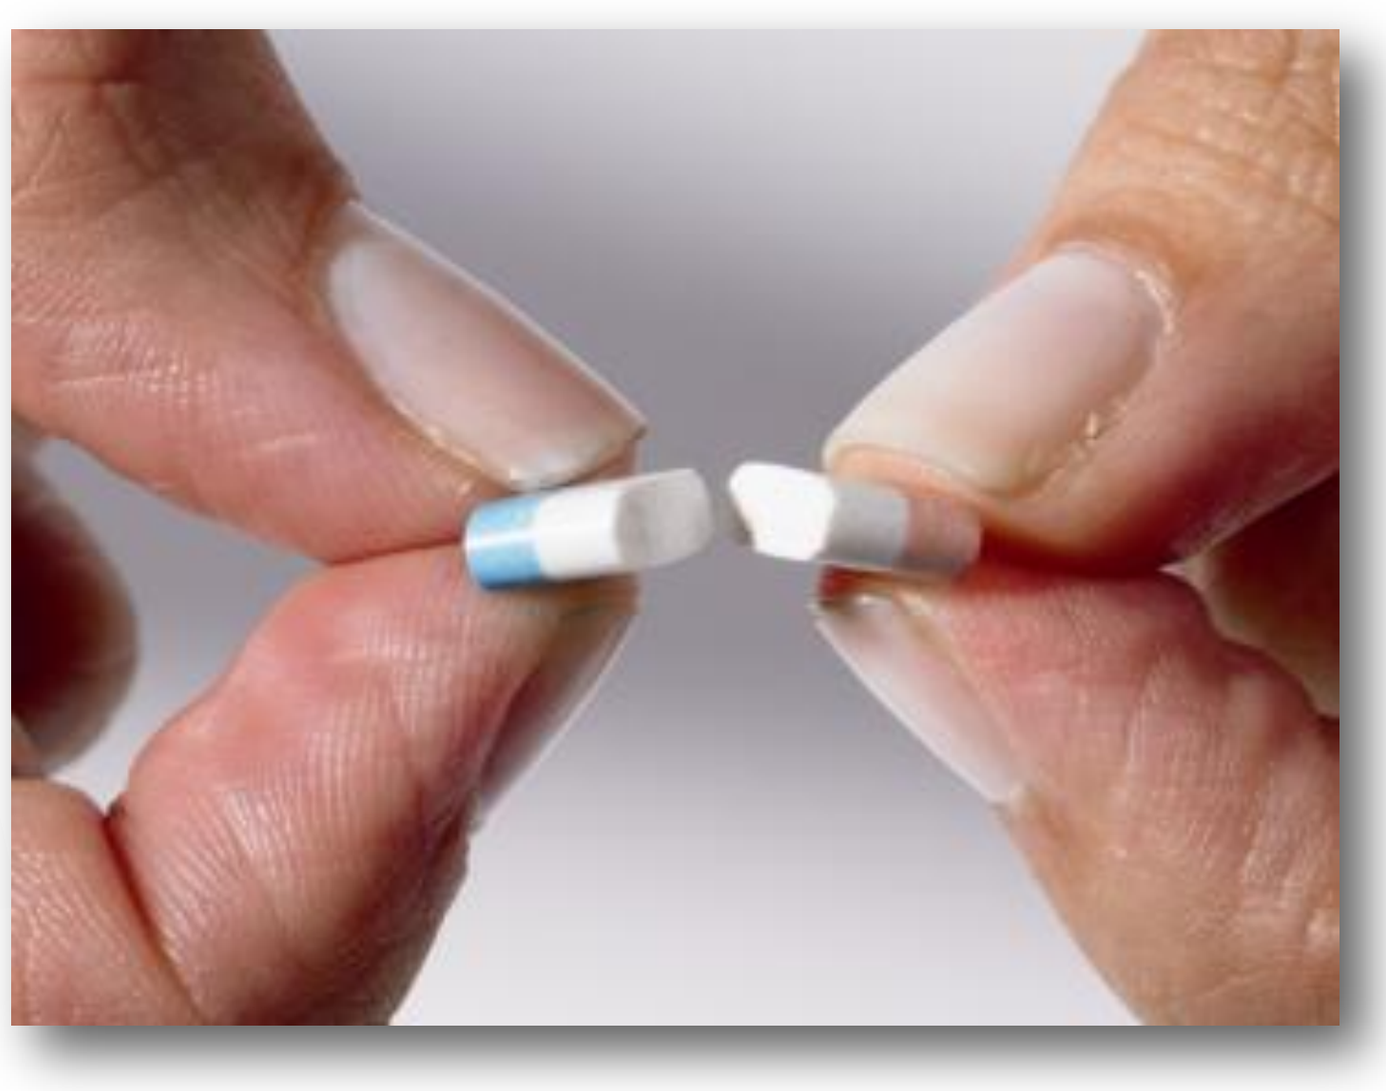

CONCLUSION

CR dosage forms offer clinical and convenience advantages for patients, but dosage form flexibility is not one of them. The nature of CR drug delivery systems makes it difficult and potentially dangerous to modify the dosage form, so nearly all CR medications are marketed as capsules or unscored tablets with instructions to not cut, crush or chew. However, the majority of solid oral CR medications on the market in the US recommend titration as part of the dosing guidelines, leaving patients with few options in the event of a change in dose. Technologies exist that allow CR formulations to be made as tablets that can be subdivided if necessary without compromising either the accuracy of the dose or the drug delivery system. This could result in better patient compliance and a more efficient use of heath care resources.

REFERENCES

1. Shah VP, Yamamoto LA, Schuirman D, Elkins J, Skelly JP. Analysis of in Vitro dissolution of whole vs. half controlled-release theophylline tablets. *Pharm Res.* 1987;4(5):416-9. 2. De Haan P, Leek CF. Oral Controlled Release Dosage Forms: A Reivew. *Pharm Weekbl Sci*. 1984;(6):57-67. 3. Primrose WR, Clee MD, Moody JP, Hockings N. Alteration of pharmacokinetics after halving a slow release theophylline tablet. *Pharmatherapeutica.* 1983;3(6): 429-432. 4. Das NG, Das SK. Controlled-Release of Oral Dosage Forms. *Formulation, Fill and Finish* (Supplement to *Pharmaceutical Technology)*. 2003;10-16. 5. Center for Drug Evaluation and Research. U.S. Food and Drug Administration website. http://www.fda.gov/cder/ob/. Accessed December 12, 2008. 6. Epocrates Inc. website. http://www.epocrates.com/. Accessed November 22, 2008. 7. Drug Information Online. Drugs.com website. http://www.drugs.com/. Accessed November 22, 2008. 8. *Physicians' Desk Reference*. 62nd ed. Montvale, NJ: Thomson PDR; 2008. 9. Sanwa Kagaku Kenkyusho Co.,Ltd. Website. www.osdrc.com. Accessed December 9, 2008.

DISCLOSURES

G.A. Green and N. Valdez are employed by Accu-Break Pharmaceuticals, Inc..
C. Berg is a Consultant for Accu-Break Pharmaceuticals, Inc.